Topical Silicone Gel for the Treatment of Surgical Scars: A Single-Center Study on Patient Satisfaction

Stephen M. Schleicher, MD

DermDOX Center for Dermatology, Hazleton, PA

Introduction

- Scars are an undesirable, yet normal outcome of wound healing[1]
- They can be conspicuous and have a significant impact on an individual's emotional and physical well-being[1]
- Several therapeutic options, both invasive and noninvasive, are available for the management of scars[1]
- Silicone-based products are recommended as the "gold standard," first-line, noninvasive treatment for linear and widespread hypertrophic scars and keloids[1-3]
- A 100% clear, medical-grade topical silicone gel is available on prescription for the management of hypertrophic and keloid scarring that results from surgery, burns, and trauma wounds
- Patient satisfaction and physician assessment with the use of this topical silicone gel for the treatment of surgical scars were evaluated in this real-world survey study

Methods

- Survey study evaluating patient satisfaction and physician's assessment with using 100% medical-grade silicone gel for 12 weeks to treat surgical scars (**Figure 1**)

Figure 1. Study design

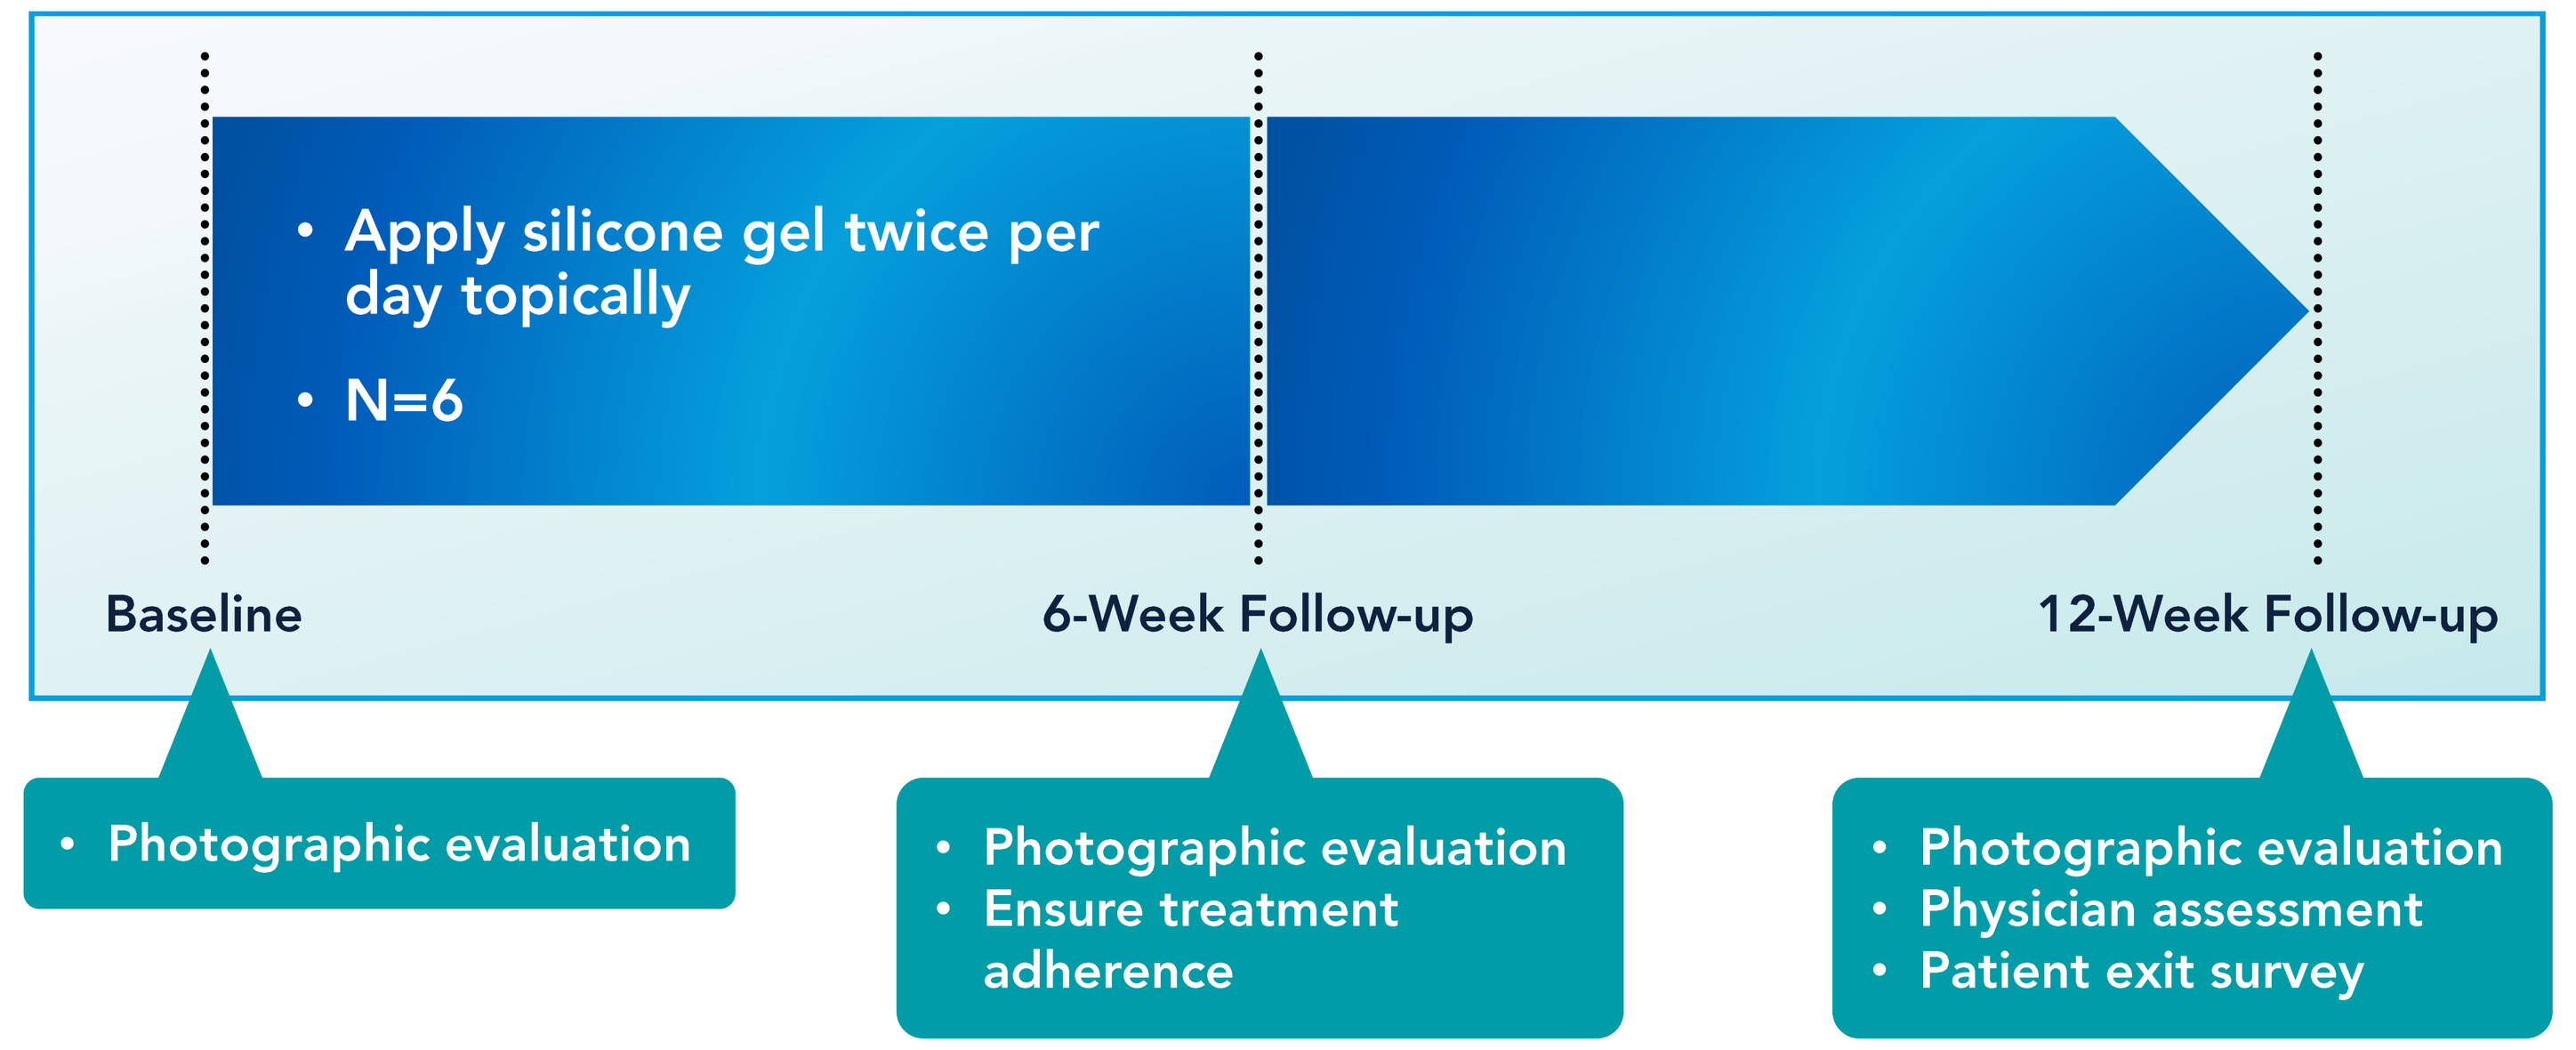
• Apply silicone gel twice per day topically
• N=6
Baseline
6-Week Follow-up
12-Week Follow-up
• Photographic evaluation
• Photographic evaluation
• Ensure treatment adherence
• Photographic evaluation
• Physician assessment
• Patient exit survey

- 6 patients (4 women, 2 men) with surgical scars were enrolled in the study
- Patients were permitted to apply the gel more frequently than twice per day, if needed
- Concomitant use of other topical medications on the scar location was prohibited
- A 7-question exit survey was administered at week 12, followed by the physician's assessment of the scar based on photographic evaluation

Results

Table 1. Baseline distribution of age, gender, and type of scar among patients enrolled in the study

Patient (Initials)	Age	Gender	Race	Type of scar
S.S.	23	Female	Caucasian	Surgical
D.V.	26	Female	Hispanic	Surgical
J.V.	40	Male	Caucasian	Surgical
B.T.	64	Female	Caucasian	Surgical
M.C.	65	Female	Caucasian	Surgical
M.H.	75	Male	Caucasian	Surgical

Patient's Evaluation of Safety and Efficacy After 12 Weeks of Treatment

- All patients reported using the topical silicone gel for the entire 12 weeks
- No itching, stinging, burning, rash, or any other side effects were reported
- The majority of patients thought that their scars had decreased in size, and had slightly and moderately softened after 12 weeks of treatment (**Figure 2**)

Figure 2. Patients reporting reduction in scar size and scar softening

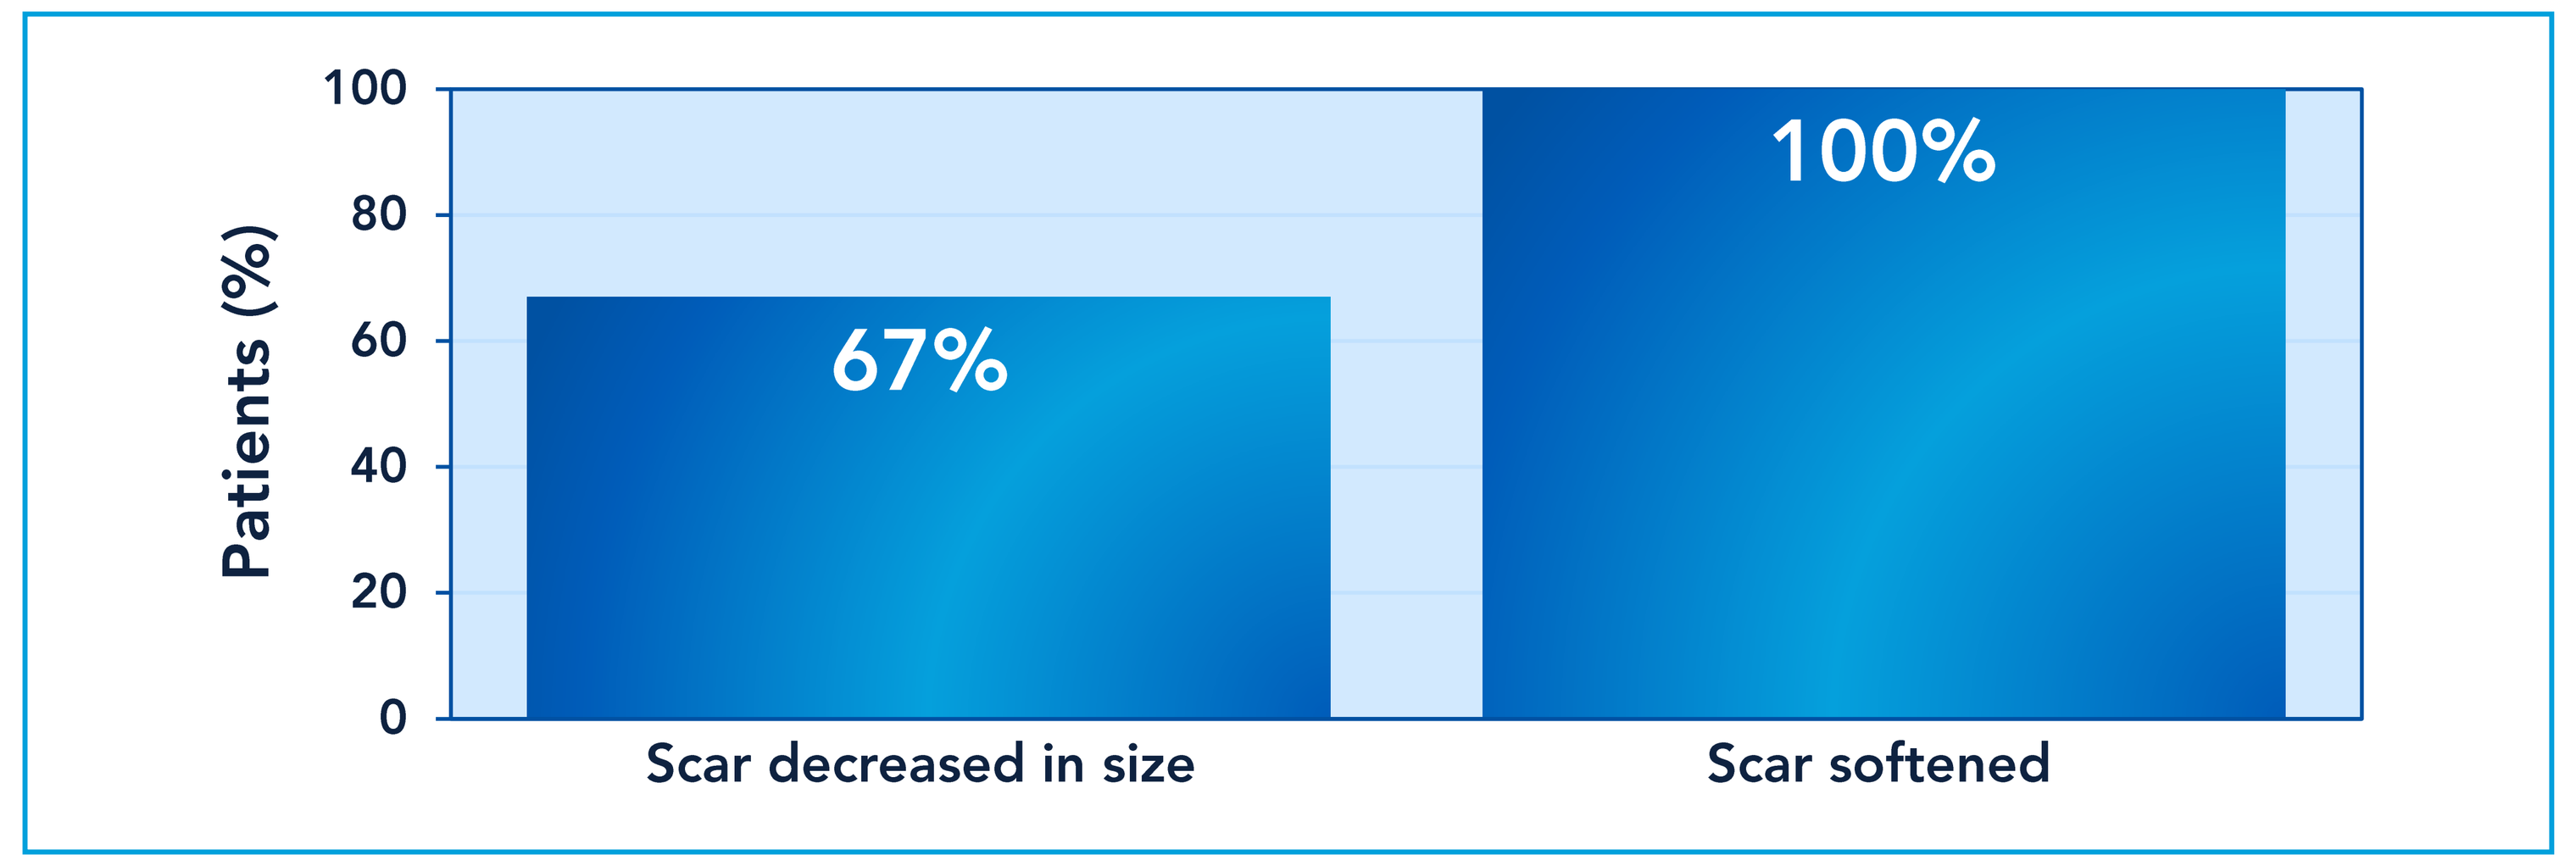
Patients (%)
100
80
60
40
20
0
67%
100%
Scar decreased in size
Scar softened

Patient Satisfaction Survey at Week 12

- Patients were generally satisfied with the use of the silicone gel at week 12 (**Figure 3**)

Figure 3. Patient satisfaction response

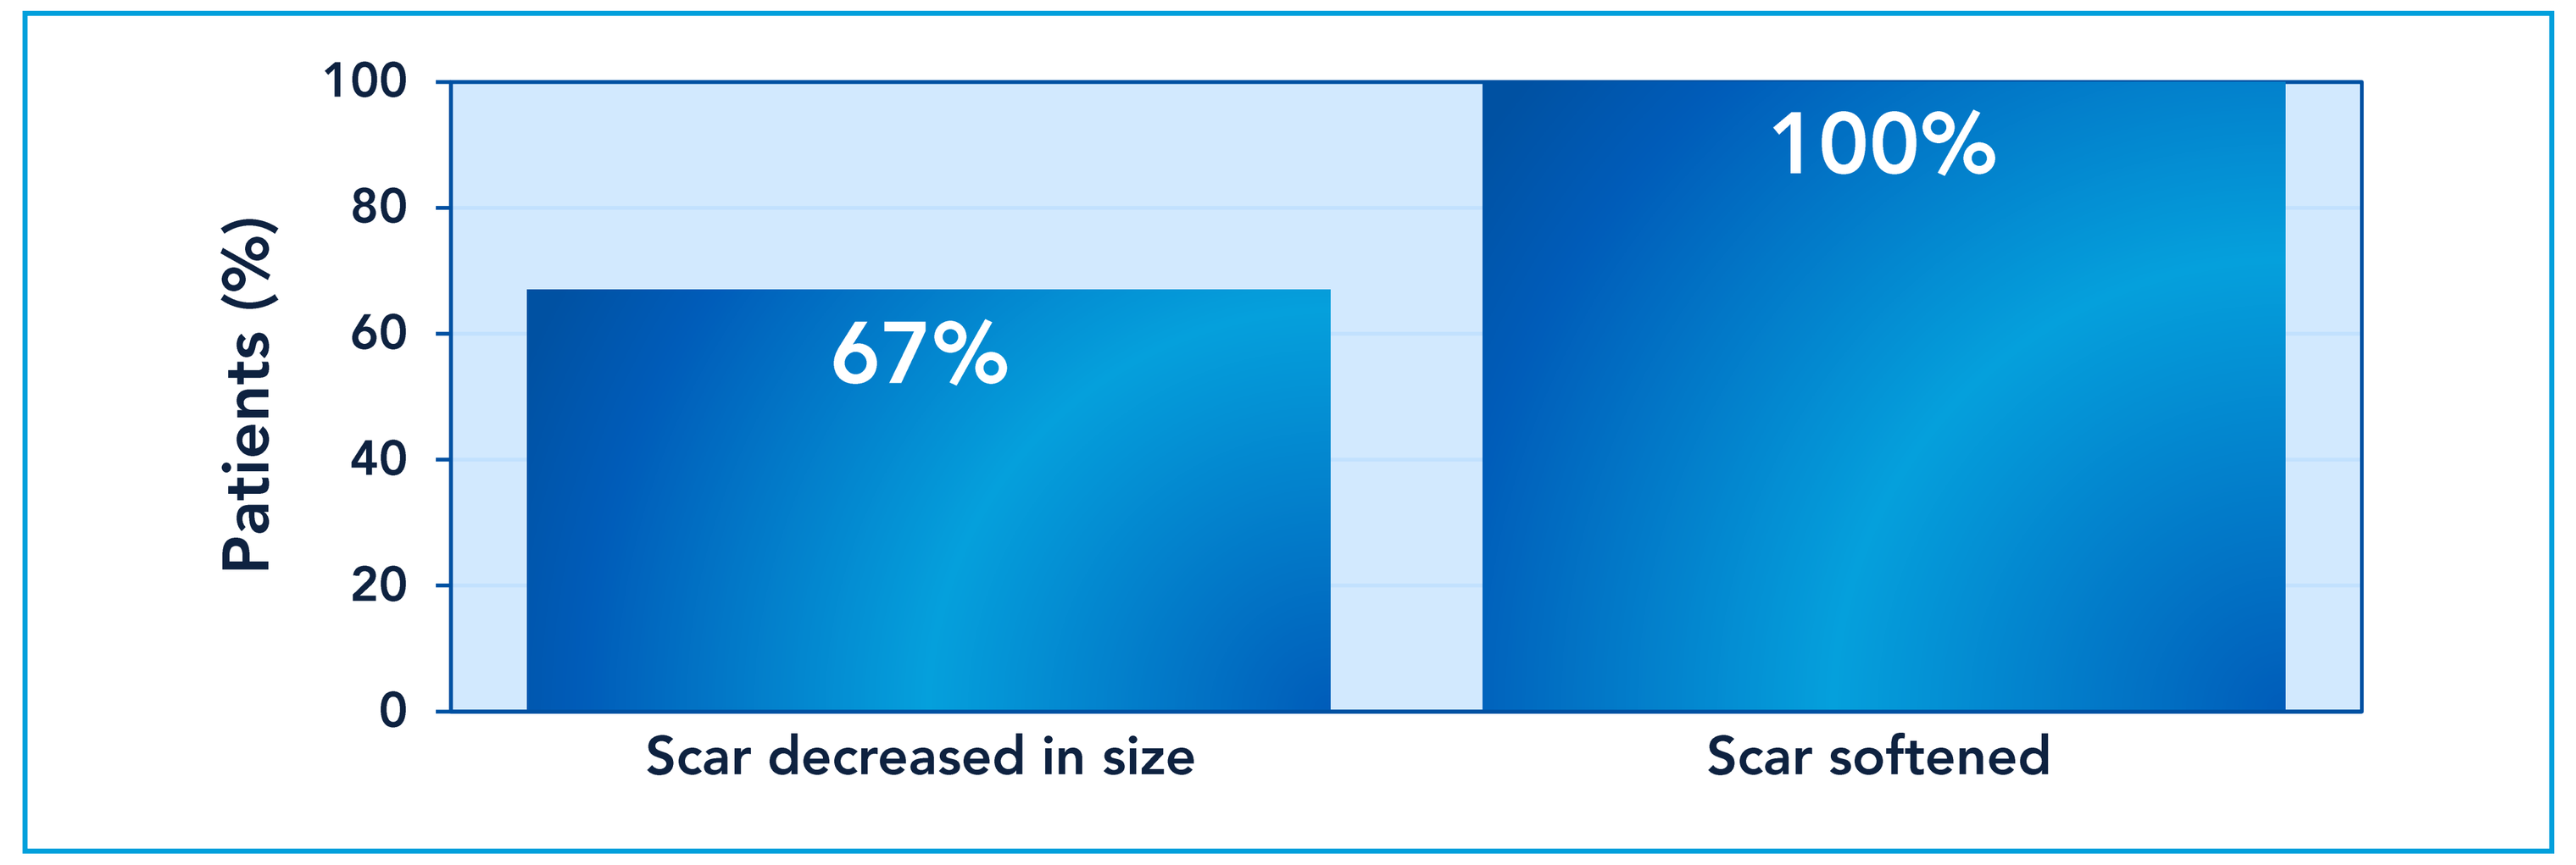
Patients (%)
100
80
60
40
20
0
67%
100%
Scar decreased in size
Scar softened

- Unsolicited written comments on the survey forms described other responses
 - "It stopped the itching and burning. The only thing that soothed me"
 - "It would not dry"

Physician Assessment and Photographic Evaluation After 12 Weeks of Treatment

- Patient D.V.'s scar showed improvement after 12 weeks of using the silicone gel (**Figure 4a**)

Figure 4a. Patient D.V. after 12 weeks of treatment

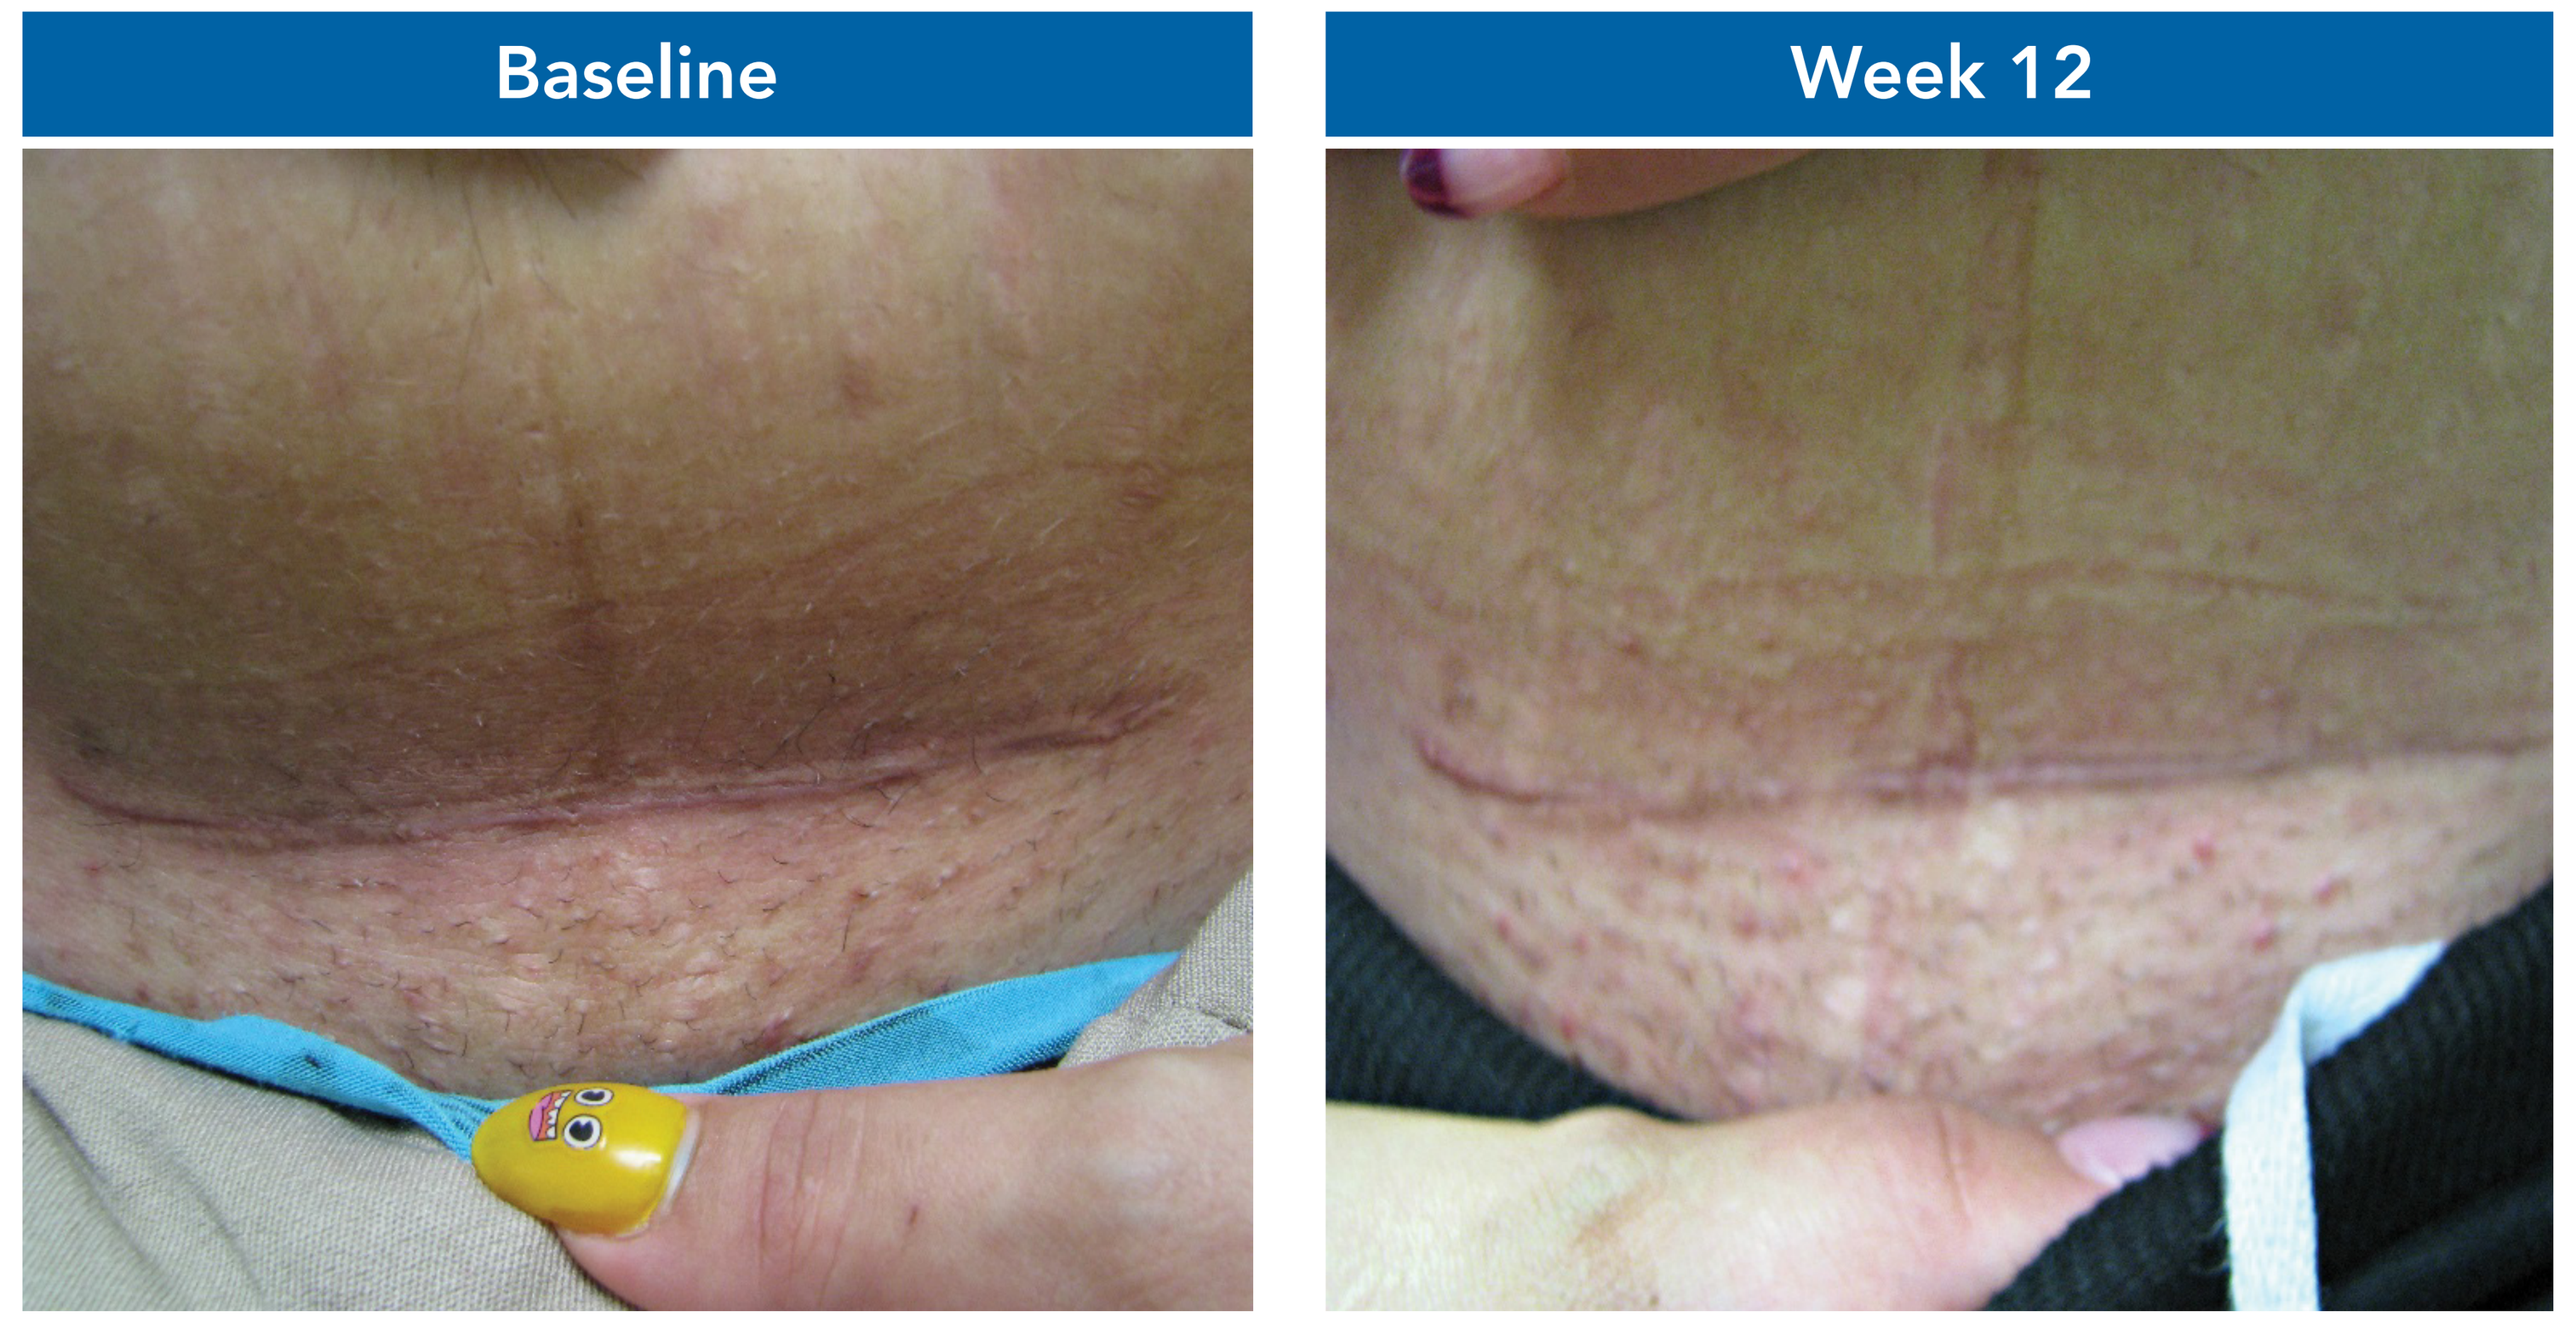
Baseline
Week 12

Physician's assessment:
Scar decreased in both elevation and diameter

- Patient M.H.'s scar showed improvement after 12 weeks of using the silicone gel (**Figure 4b**)

Figure 4b. Patient M.H. after 12 weeks of treatment

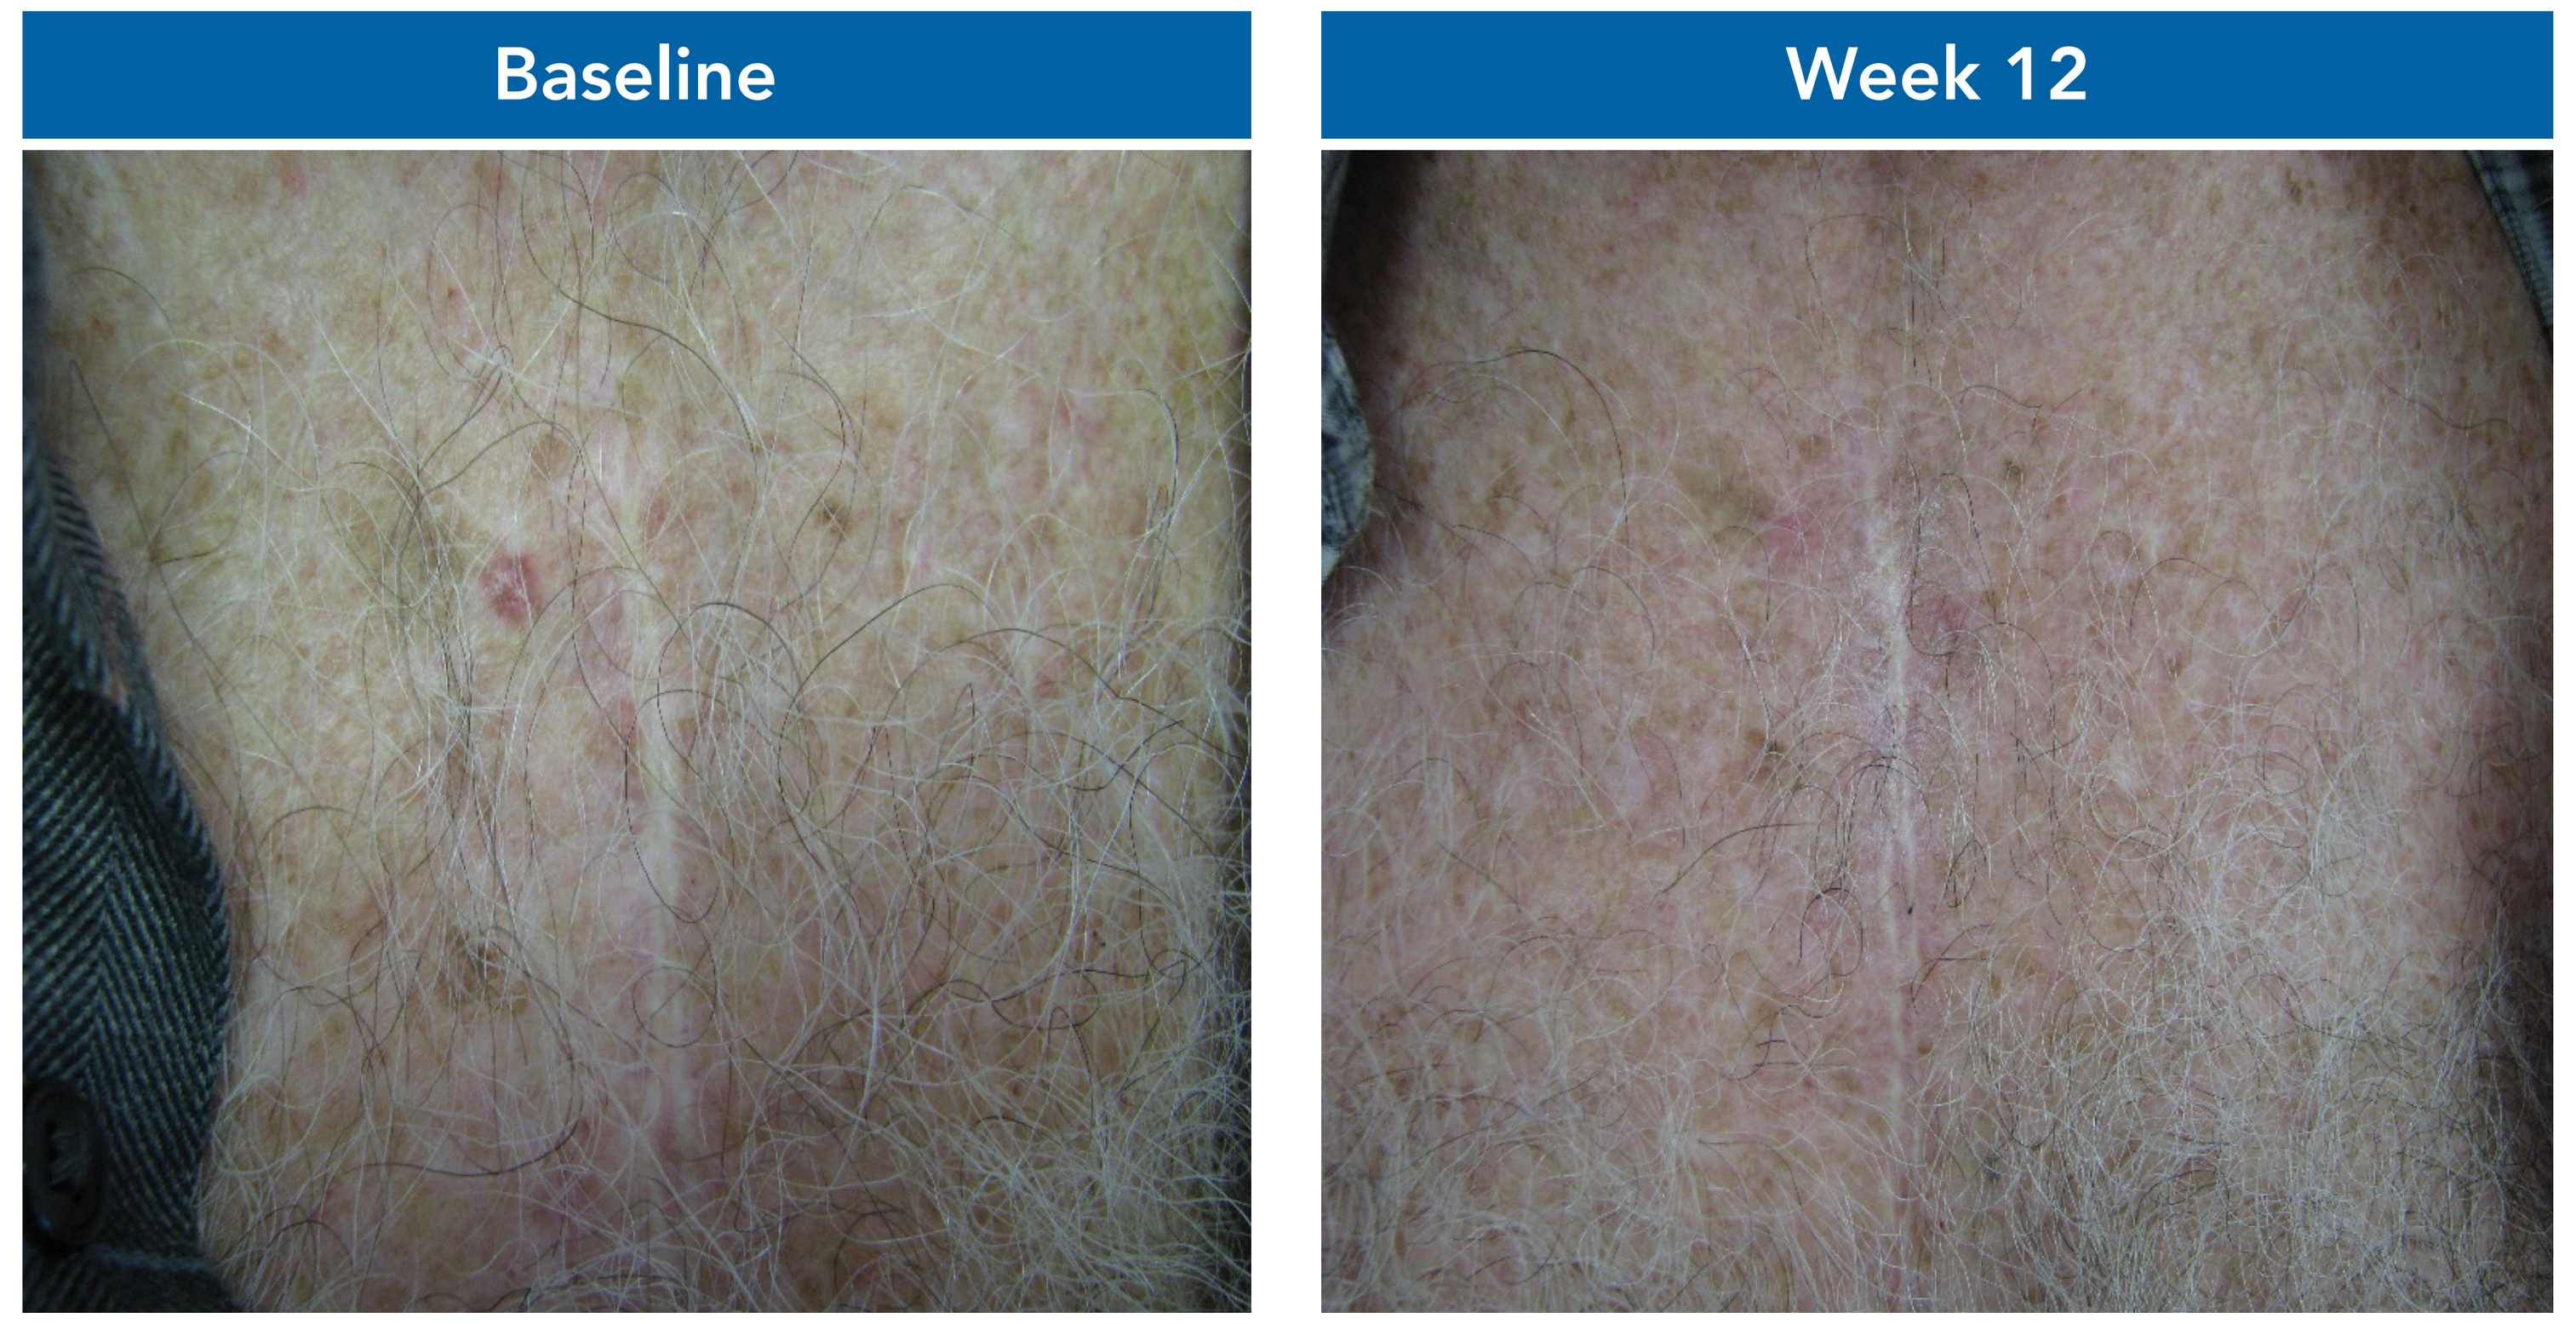
Baseline
Week 12

Physician's assessment:
Scar decreased in diameter

Figure 4c. Patient B.T. after 12 weeks of treatment

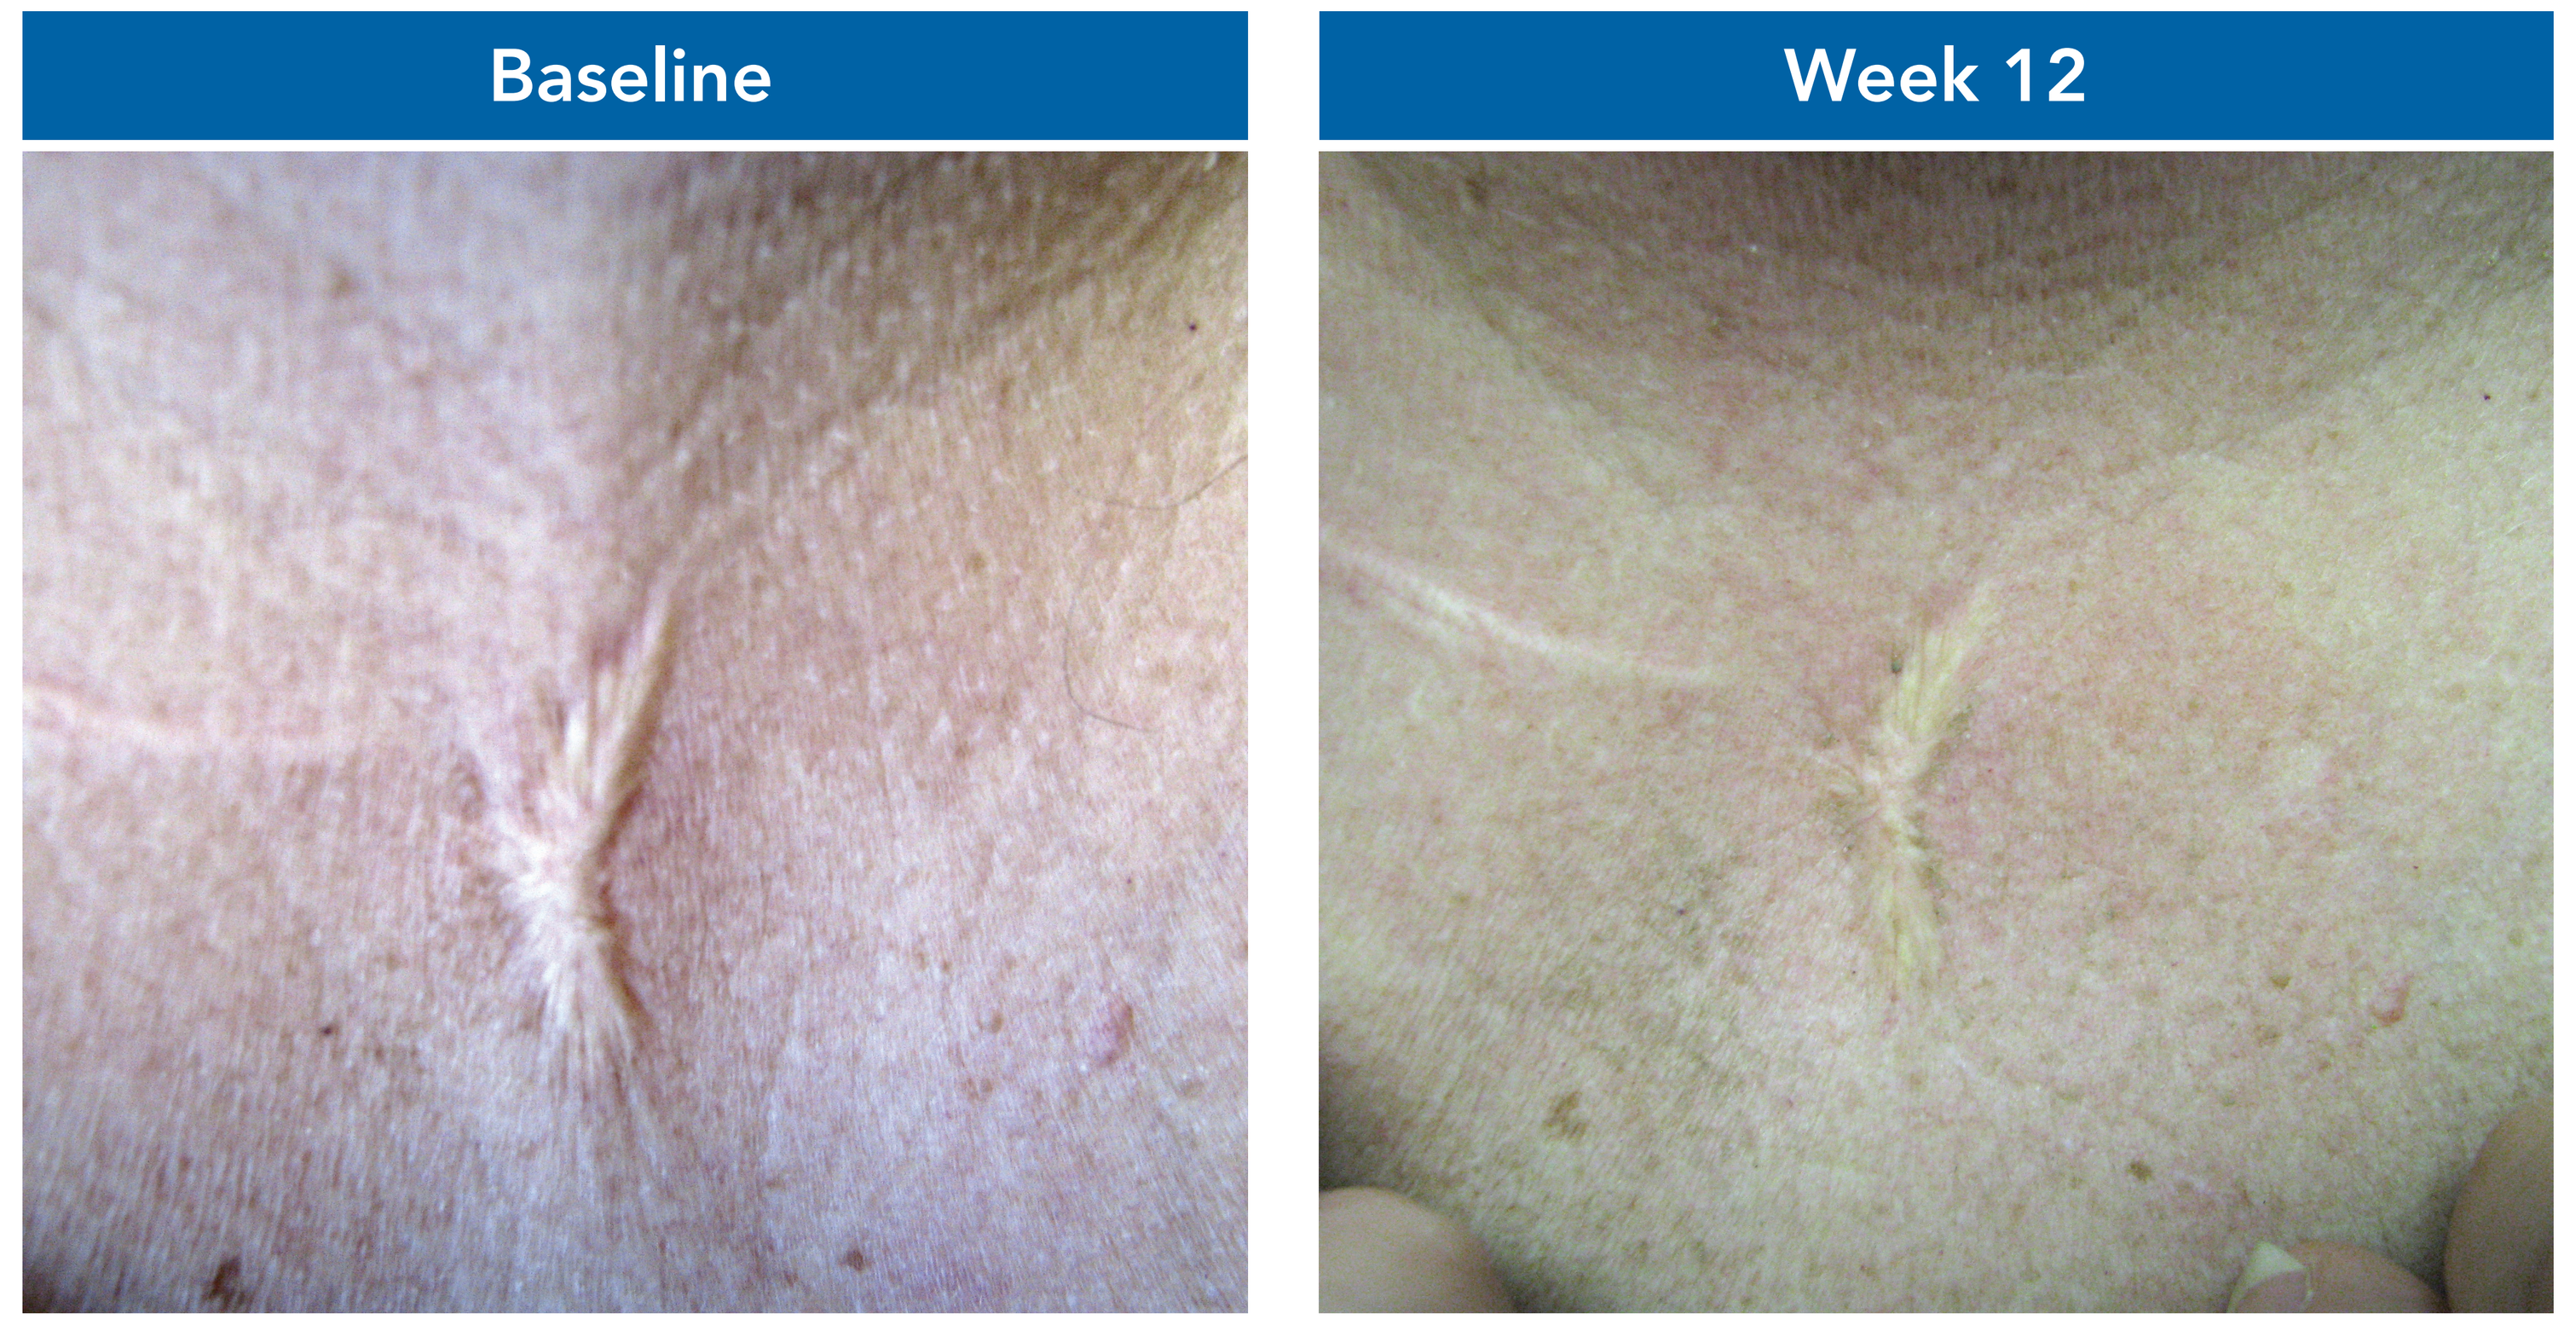
Baseline
Week 12

- Physician's assessment indicated that no scar worsened for the other 4 patients at week 12

Conclusions

- Patients were highly satisfied with the use of the 100% medical-grade topical silicone gel to treat surgical scars
- There were no reported adverse reactions after 12 weeks of treatment
- All patients reported softening of their scars (rated as slightly or moderately), and the majority thought that their scars had decreased in size after 12 weeks of treatment with the silicone gel
- Physician assessment indicated scar improvement (decreases in diameter and/or elevation) in 2 patients and no worsening of scars in the remaining cohort
- The majority of patients found the gel easy to apply and liked the feeling on the skin
 - One patient indicated that this 100% medical-grade silicone gel was the only product that soothed the itching and burning feeling

References:
1. Meaume S, et al. *Eur J Dermatol.* 2014;24:435-443.
2. Gold MH, et al. *Dermatol Surg.* 2014;40:825-831.
3. Monstrey S, et al. *J Plast Reconstr Aesthet Surg.* 2014;67:1017-1025.

Supported by a grant from Exeltis USA.